Evaluation of the Physical and Chemical Compatibility of Tacrolimus Ointment, 0.1% (Fujisawa) and Desoximetasone Ointment, 0.25% (Taro)

By Jacob Levitt, M.D., Terry Feldman, Ph.D.**, Ildiko Riss, M.Sc.**, and On-Tai Leung, Ph.D., Cchem.***

Abstract

Desoximetasone ointment, 0.25% and tacrolimus ointment, 0.1% are widely used in the treatment of atopic dermatitis. To determine if these drugs are physically and chemically compatible when mixed, a one-to-one mixture of desoximetasone ointment, 0.25% (Taro) and Protopic® ointment, 0.1% (Fujisawa) was prepared and stored under three different temperature/humidity conditions. Unmixed ointments served as controls. Mixed and unmixed samples were stored at 25ºC, 60% relative humidity, 30ºC, 60% relative humidity, and 40ºC, 75% relative humidity. Samples were evaluated at Days 1, 2, 7, 14, and 28 for color, degree of physical separation, and chemical stability via reverse-phase high performance liquid chromatography. Quantitation was based on authentic drug substance standard for desoximetasone and was based on Protopic Injection® (5mg/1mL) for tacrolimus. Mixtures remained white for all time periods except at Day 28, at which time both the desoximetasone control and the mixtures at all storage conditions were pale yellow. No more than slight separation was observed at any time period under any of the three storage conditions. For all time periods and storage conditions, potency recovery ranges for the active drug substances were as follows: Tacrolimus, Mixture: 70.0-79.2%, Tacrolimus, Control: 70.8-79.3%, Desoximetasone, Mixture: 95.2-102.9%, and Desoximetasone, Control: 93.9-103.5%. The range of % relative recovery for all storage conditions (% Mixture/% Control) x 100, are: Tacrolimus: 89.6 - 109.3% and Desoximetasone: 99.0 - 103.4%. No significant difference in either physical appearance or chromatographic profile between the mixture and individual ointments was observed. **Conclusion**: Desoximetasone ointment, 0.25% (Taro) and Protopic® ointment, 0.1% (Fujisawa) are compatible both physically and chemically up to four weeks when mixed at a ratio of 1:1 (w/w).

Objective

- To determine the physical and chemical compatibility both qualitatively and quantitatively of the combination of tacrolimus 0.1% ointment (Protopic®) and desoximetasone 0.25% ointment (generic of Topicort®)

Significance

- Doctors prescribe Protopic® and steroids in atopic dermatitis, psoriasis, and seborrheic dermatitis.
- Desoximetasone 0.25% ointment is a Group II steroid
 – Side effects: atrophy, telangiectasia, hypopigmentation
- Protopic® 0.1% ointment is a non-steroid topical immunomodulator
 – Side effects: itching, burning, stinging
- Physical compatibility allows confident SIMULTANEOUS application, theoretically decreasing tacrolimus side effects and enabling further steroid-sparing
- Some compounds are not stable when mixed, due to:
 – Compound/compound incompatibility
 – Vehicle/compound incompatibility
 – Vehicle/vehicle incompatibility

Examples of Incompatibility

- **Dovonex® 0.005% ointment** and:
 – Salicylic acid 6% ointment
 – Ammonium lactate 12% lotion
 – Hydrocortisone-17-valerate 0.2% ointment
- **Tazarotene 0.05% gel** and:
 – Betamethasone dipropionate 0.05% gel (vs. compatible with ointment, cream, or lotion)
 – Clobetasol propionate 0.05% gel (vs. compatible with ointment)

Chemical Structures

Desoximetasone

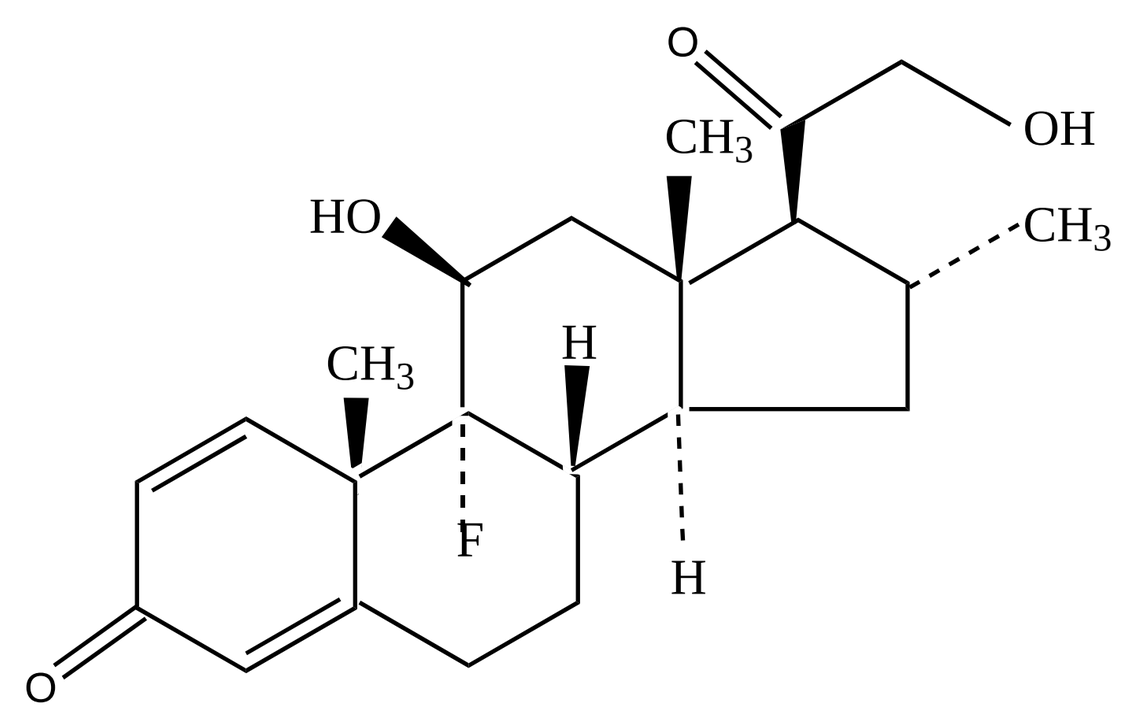

Tacrolimus

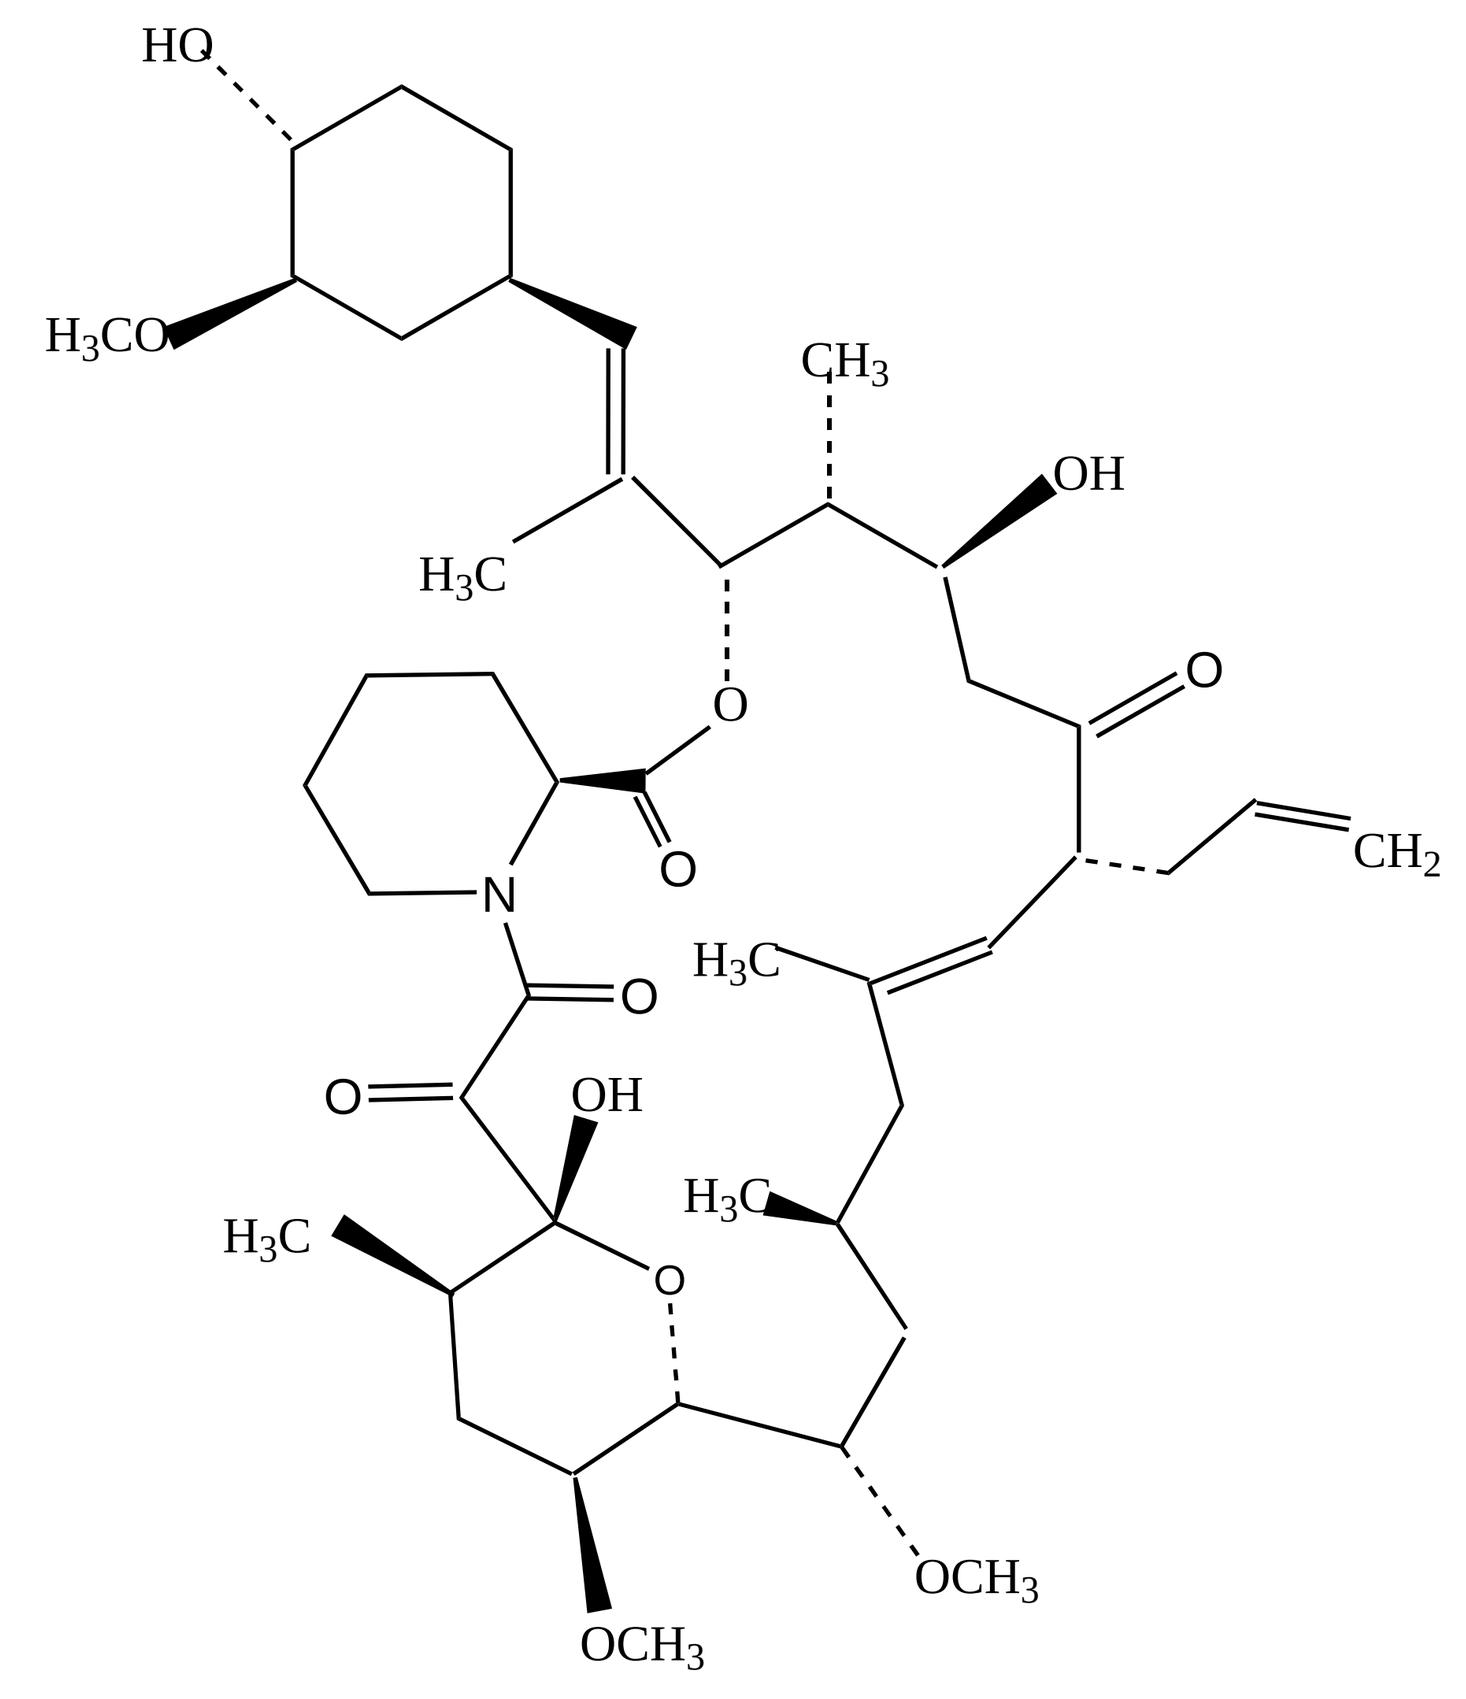

Methods

- 80g desoximetasone ointment 0.25% (Taro) and 80g Protopic® ointment 0.1% (Fujisawa) were mixed at room temperature and stored in glass vials (1:1 w/w)
- Stability of 10g of mixture and control (non-mixed) samples were measured at **Day 1, 2, 7, 14, and 28** under three different temperature/humidity conditions:
 - **25ºC, 60% relative humidity**
 - **30ºC, 60% relative humidity**
 - **40ºC, 75% relative humidity**

- Compatibility was measured by:
 - color and degree of physical separation, determined visually
 - degree of degradation, determined by HPLC
 – based on authentic drug substance standard for the desoximetasone and on Protopic Injection® (5mg/1mL) for tacrolimus

Results – Qualitative

- No more than slight separation was observed over any time period under any of the three storage conditions
- Mixtures remained white, except for pale yellow appearance at Day 28 under all storage conditions of the mixture and desoximetasone control

Results – Quantitative

For all times and storage conditions, ranges of recovered materials were:
- Tacrolimus, Mixture: 70.0-82.3%
- Tacrolimus, Control: 67.9-81.3%
- Desoximetasone, Mixture: 95.2-102.9%
- Desoximetasone, Control: 93.9-103.5%

Results – Quantitative

- Range of **% Relative Recovery** (% Mixture/% Control) x 100 for All Storage Conditions and All Time Periods:
- ***Tacrolimus*: 89.5 - 109.7%**
- ***Desoximetasone*: 99.0 - 103.4%**

Graphically

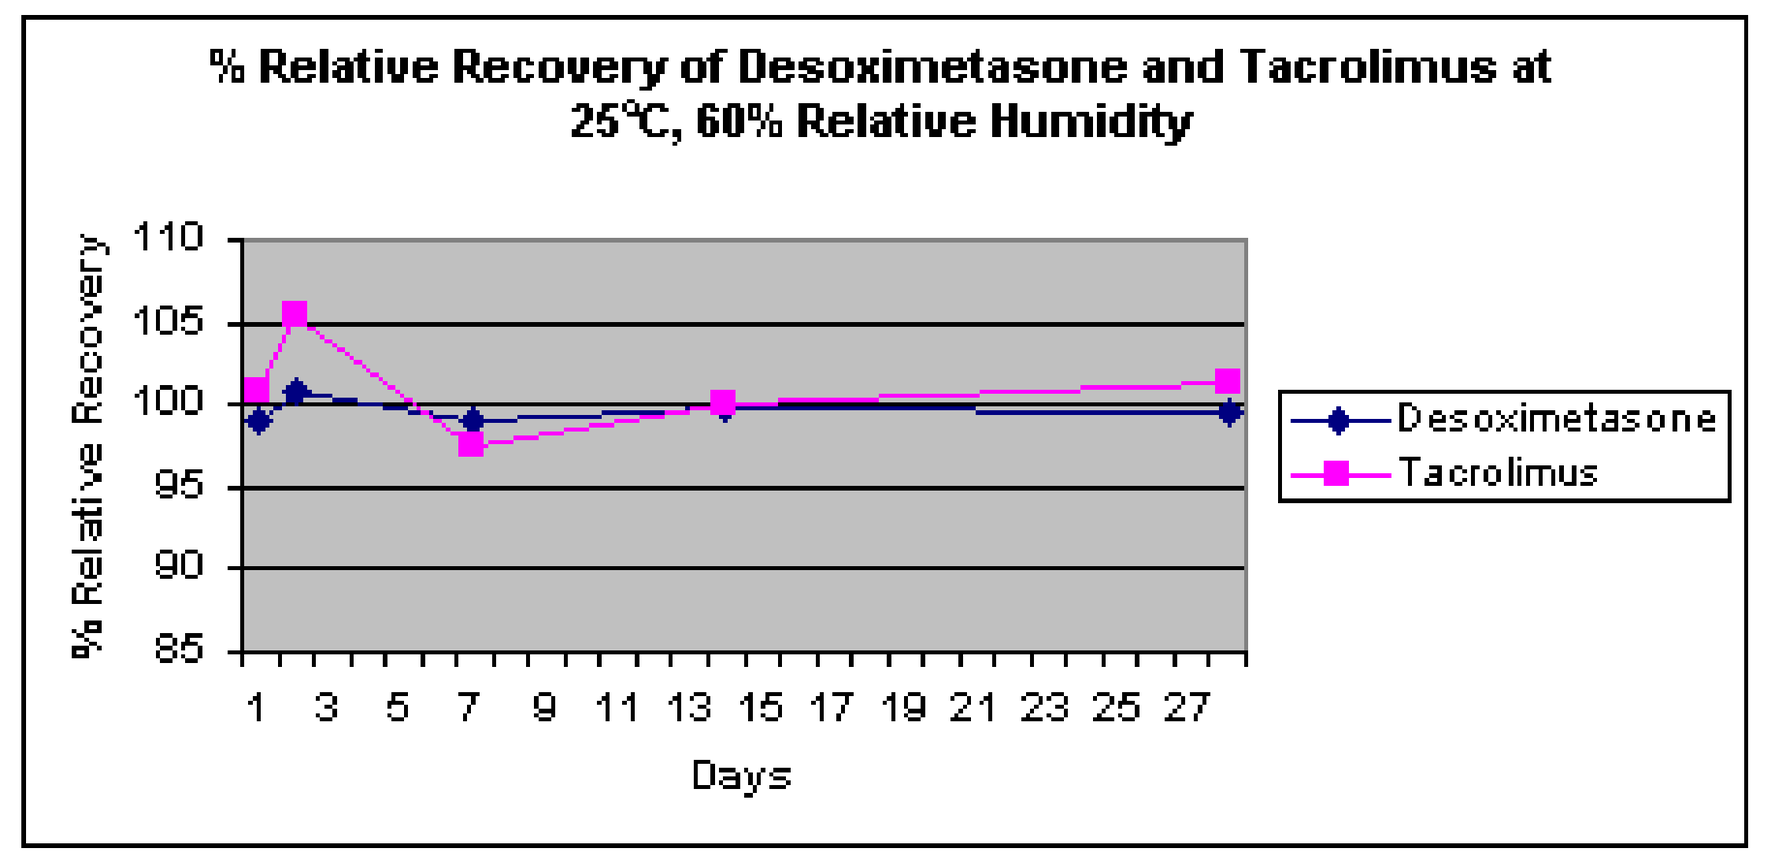

% Relative Recovery of Desoximetasone and Tacrolimus at 25°C, 60% Relative Humidity
% Relative Recovery
110
105
100
95
90
85
1 3 5 7 9 11 13 15 17 19 21 23 25 27
Days
Desoximetasone
Tacrolimus

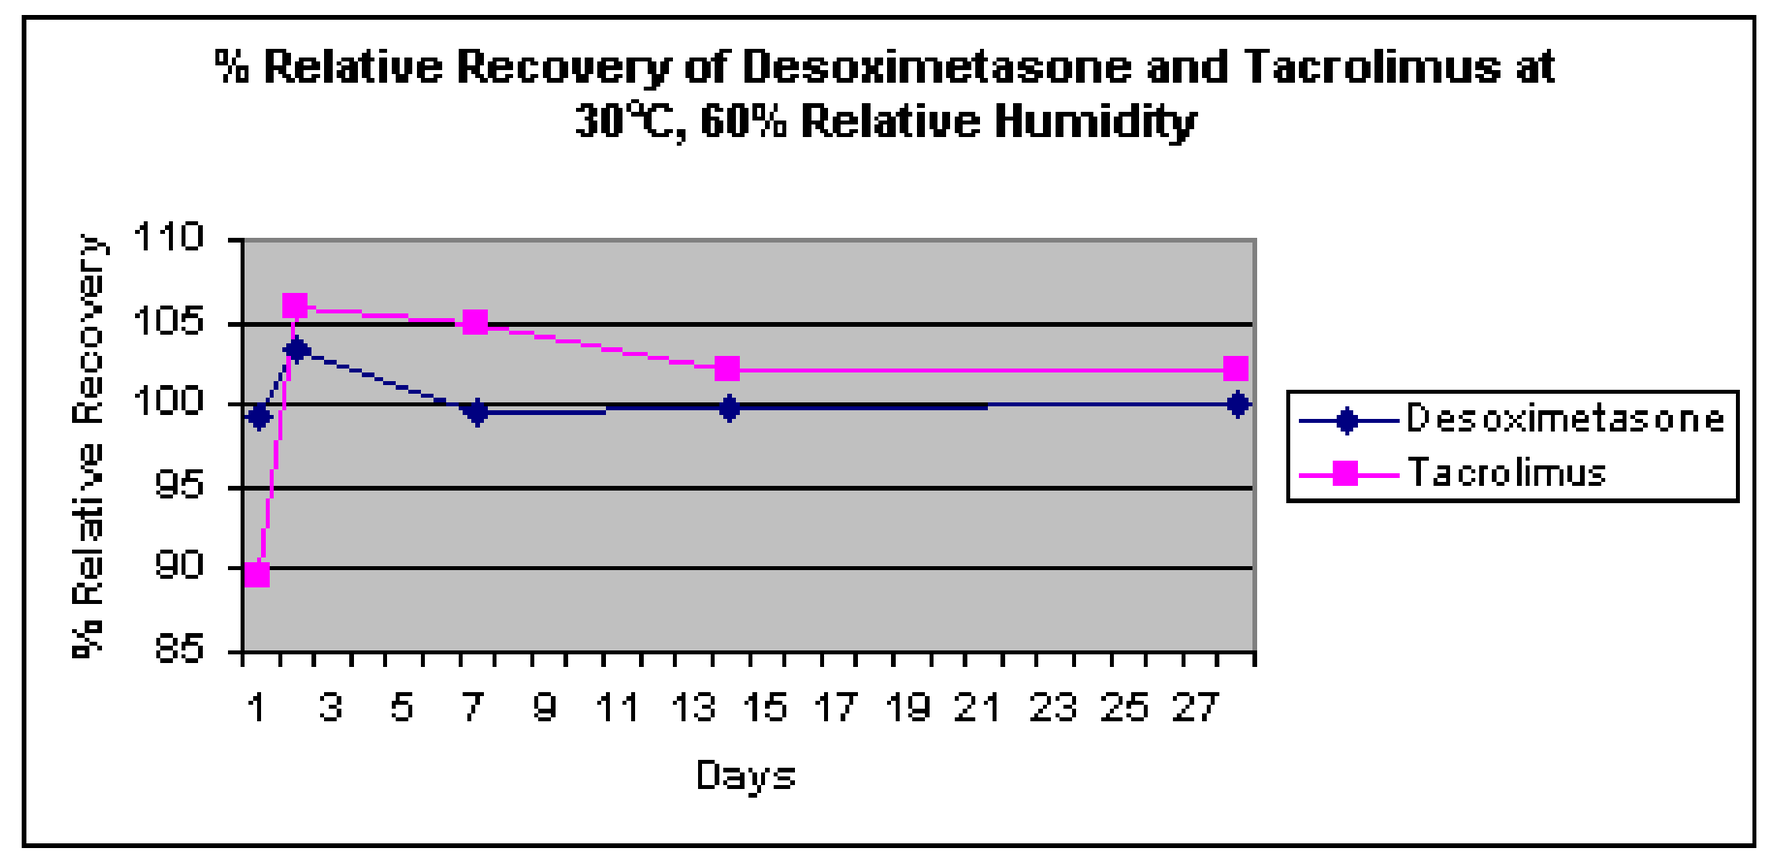

% Relative Recovery of Desoximetasone and Tacrolimus at 30°C, 60% Relative Humidity
% Relative Recovery
110
105
100
95
90
85
1 3 5 7 9 11 13 15 17 19 21 23 25 27
Days
Desoximetasone
Tacrolimus

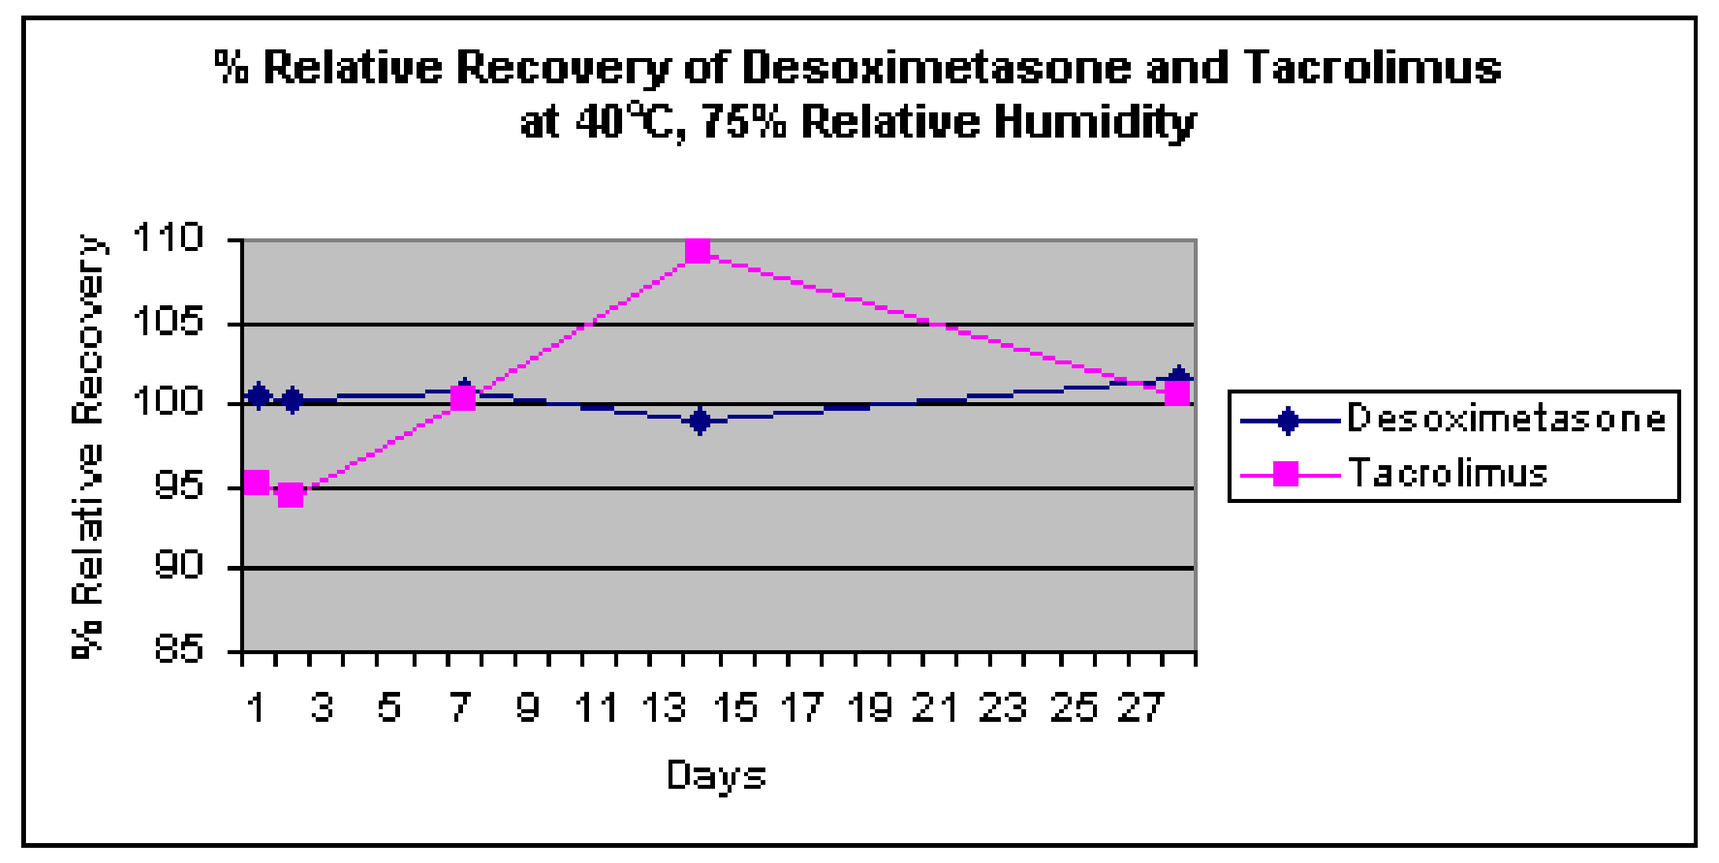

% Relative Recovery of Desoximetasone and Tacrolimus at 40°C, 75% Relative Humidity
% Relative Recovery
110
105
100
95
90
85
1 3 5 7 9 11 13 15 17 19 21 23 25 27
Days
Desoximetasone
Tacrolimus

Conclusion

- There is *no significant difference* in either physical appearance or chromatographic profile between the mixture and individual ointments.
- **Protopic ointment, 0.1% (Fujisawa) and desoximetasone ointment, 0.25% (Taro) are compatible both physically and chemically up to 4 weeks when mixed at a ratio of 1:1 (w/w).**

References

- Hecker, D, Worsley, J, Yueh, G, Lebwohl, M. In vivtro compatibility of tazarotene with other topical treatments of psoriasis. *J Am Acad Dermatol* 2000; 42(6):1008-11.
- Patel, B, Siskin, S, Krazmien, R, Lebwohl, M. Compatibility of capcipotriene with other topical medications. *J Am Acad Dermatol* 1998; 38(6):1010-1.
- Nghiem, P, Pearson, G, Langley, RG. Tacrolimus and pimecrolimus: From clever prokaryotes to inhibiting calcineurin and treating atopic dermatitis. *J Am Acad Dermatol* 2002; 46:228-41.
- Namiki, Y, Kirhara, N, Koda, S, Hane, K, Yasuda, T. Tautomeric phenomenon of a novel potent immunosuppressant (FK506) in solution. I. Isolation and structure determination of tautomeric compounds. *The Journal of Antibiotics* 1993; 46(7):1149-1155.
- Akashi, T, Nefuji, T, Yoshida, M, Hosoda, J. Quantitative determination of tautomeric FK506 by reversed-phase liquid chromatography. *J of Pharmaceutical and Biomedical Analysis* 1996; 14:339-346.
- Nishikawa, T, Hasumi, H, Suzuki, S, Kubo, H, Ohtani, H. Kinetic analysis of molecular interconversion of immunosuppressant FK506 by high-performance liquid chromatography. *Pharmaceutical Research* 1993; 10(12):1785-1789.
- Jacobson, PA, Johnson, CE, West, NJ, Foster, JA. Stability of tacrolimus in an extemporaneously compounded oral liquid. *American Journal of Health-System Pharmacy* 1997; 54(2):178-180.

*Department of Dermatology, Mount Sinai Medical Center, New York, NY 10029
**Taro Pharmaceuticals Inc., 130 East Drive, Brampton, Ontario L6T 1C1